Presented at:
San Antonio Breast Cancer Symposium
December 6-10, 2022
Poster # OT2-01-03

ELEVATE: A Phase 1b/2, Open-Label, Umbrella Study Evaluating Elacestrant in Various Combinations in Women and Men With Metastatic Breast Cancer (mBC)

Hope S. Rugo,[1] Aditya Bardia,[2] Javier Cortes,[3] Giuseppe Curigliano,[4,5] Erika Hamilton,[6] Sara Hurvitz,[7] Sibylle Loibl,[8] Simona Scartoni,[9] Tarek Sahmoud,[10] Krzysztof J. Grzegorzewski,[10] Nassir Habboubi,[10] Joyce O'Shaughnessy[11]

[1]University of California San Francisco Comprehensive Cancer Center, San Francisco, CA, USA; [2]Massachusetts General Hospital Cancer Center, Harvard Medical School, Boston, MA, USA; [3]International Breast Cancer Center (IBCC), Quiron Group, Barcelona, Spain; [4]European Institute of Oncology IRCCS, Milan, Italy; [5]University of Milano, Milan, Italy; [6]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN, USA; [7]UCLA Jonsson Comprehensive Cancer Center, Los Angeles, CA, USA; [8]German Breast Group, Neu-Isenburg, Germany; Centre for Haematology and Oncology Bethanien, Frankfurt, Germany; [9]Menarini Group, Florence, Italy; [10]Stemline Therapeutics/Menarini Group, New York, NY, USA; [11]Baylor University Medical Center, Texas Oncology, US Oncology, Dallas, TX, USA

BACKGROUND

- Estrogen receptor-positive (ER+) and HER2-negative (HER2-) breast cancer is the most prevalent breast cancer subtype. Endocrine therapy is the mainstay of treatment; however, due to the heterogeneous nature of the disease, development of resistance to this therapeutic approach is very common in the metastatic setting.[1]
- After progression on first-line treatment, both hormonal monotherapy and combination therapy are available.
- Elacestrant is a novel, oral selective ER degrader that is being developed for the treatment of ER+ advanced/mBC.
- Elacestrant demonstrated significantly prolonged progression-free survival and a manageable safety profile compared with standard-of-care endocrine therapy in the phase 3 EMERALD trial, which enrolled patients with ER+ HER2– mBC following disease progression on prior endocrine and cyclin-dependent kinase 4/6 inhibitor (CDK4/6i) therapy. Benefit was observed in the overall population and in patients with ESR1 mutations.[2]
- Combining elacestrant with targeted agents utilized in combination with endocrine therapy in mBC is of therapeutic interest.

OBJECTIVES

Primary

Phase 1b
- Determine the RP2D of elacestrant in combination with each of the other study drugs
- Characterize the safety of elacestrant in combination with each of the other study drugs

Phase 2
- Evaluate the efficacy of elacestrant in combination with each of the other drugs for PFS

Secondary

Phase 1b
- Describe the plasma PK of elacestrant in combination with each of the other study drugs
- Evaluate the efficacy of elacestrant in combination with each of the other study drugs

Phase 2
- Evaluate the efficacy of elacestrant in combination with each of the other drugs for additional efficacy endpoints
- Further characterize the safety of elacestrant in combination with each of the other study drugs

Exploratory

Phase 1b and Phase 2
- Assess the PD of each combination
- Evaluate the relationship between efficacy and PD
- Evaluate the efficacy of each combination according to *ESR1* mutation status at baseline

CBR, clinical benefit rate; OS, overall survival; PD, pharmacodynamics; PFS, progression-free survival; PK, pharmacokinetics; RP2D, recommended phase 2 dose.

METHODS - STUDY DESIGN

Multicenter, Phase 1b/2 trial evaluating the RP2D of elacestrant when administered in combination with other study drugs (Phase 1b) and to evaluate the safety and efficacy of the various combinations in patients with ER+/HER2-advanced/metastatic breast cancer (Phase 2)

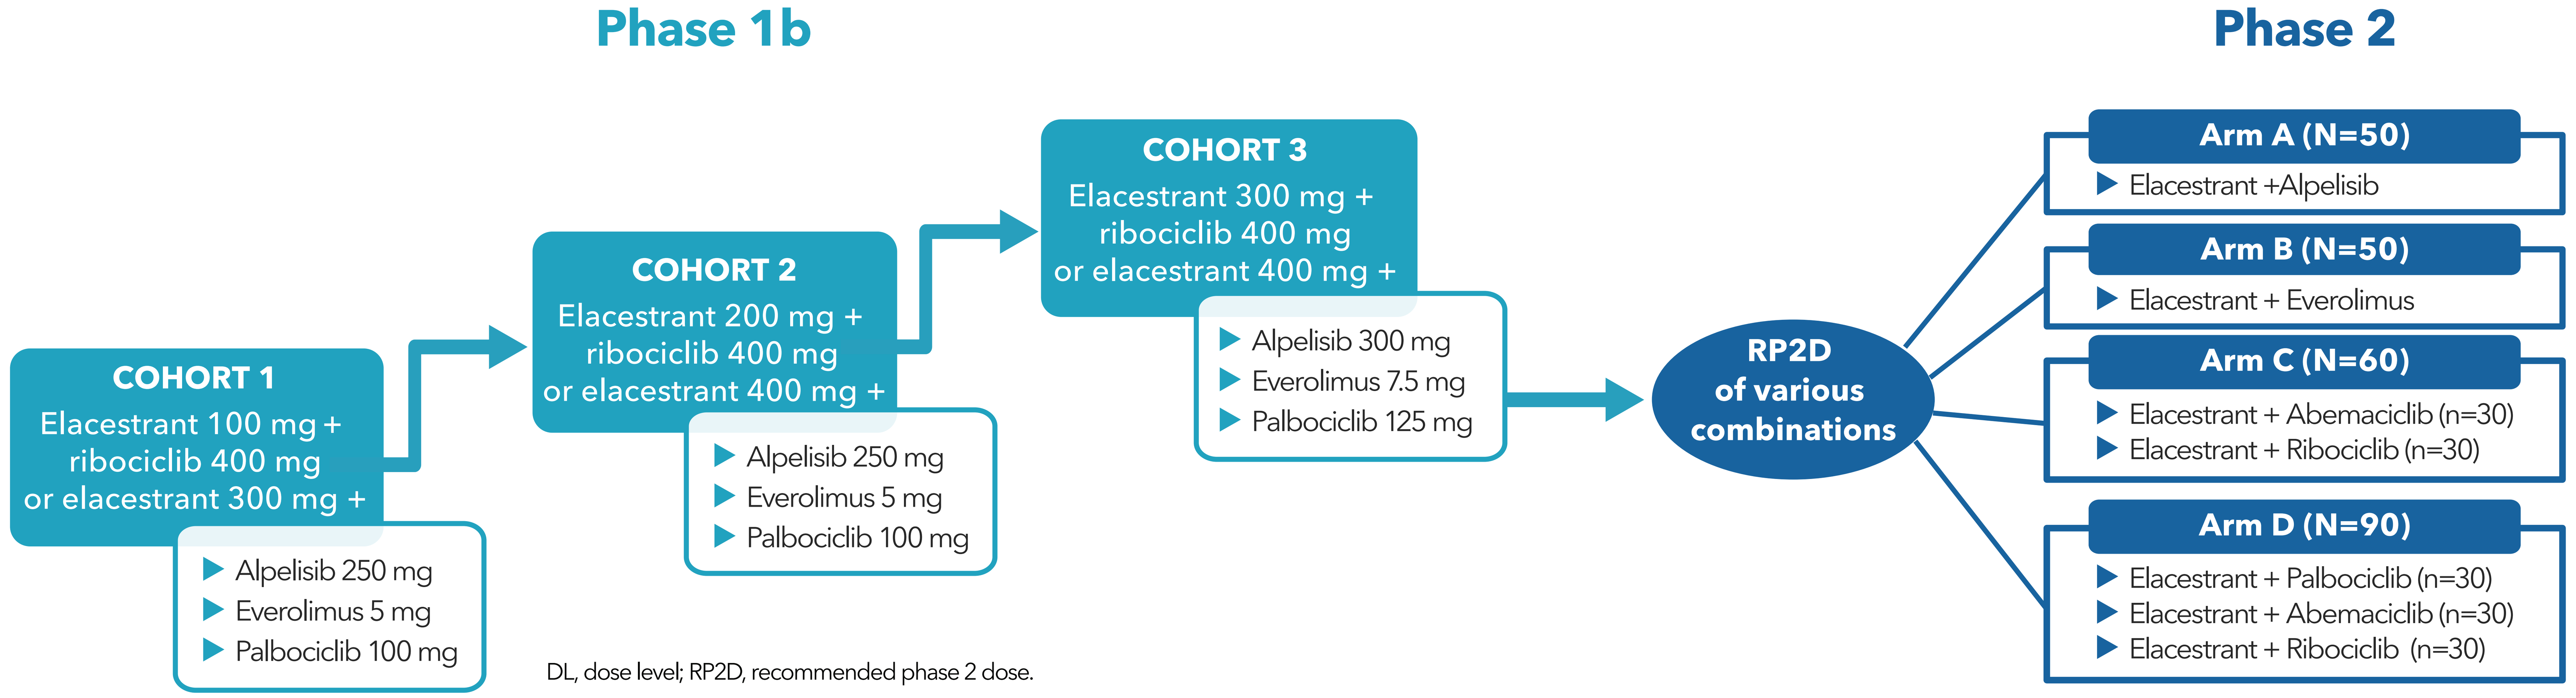
Phase 1b
Phase 2
COHORT 1
Elacestrant 100 mg + ribociclib 400 mg or elacestrant 300 mg +
Alpelisib 250 mg
Everolimus 5 mg
Palbociclib 100 mg
COHORT 2
Elacestrant 200 mg + ribociclib 400 mg or elacestrant 400 mg +
Alpelisib 250 mg
Everolimus 5 mg
Palbociclib 100 mg
COHORT 3
Elacestrant 300 mg + ribociclib 400 mg or elacestrant 400 mg +
Alpelisib 300 mg
Everolimus 7.5 mg
Palbociclib 125 mg
RP2D of various combinations
Arm A (N=50)
Elacestrant +Alpelisib
Arm B (N=50)
Elacestrant + Everolimus
Arm C (N=60)
Elacestrant + Abemaciclib (n=30)
Elacestrant + Ribociclib (n=30)
Arm D (N=90)
Elacestrant + Palbociclib (n=30)
Elacestrant + Abemaciclib (n=30)
Elacestrant + Ribociclib (n=30)

DL, dose level; RP2D, recommended phase 2 dose.

INCLUSION/EXCLUSION CRITERIA

Key Inclusion Criteria Phase 1b and Phase 2

- Women or men age ≥18 years
- Female patients may be postmenopausal, premenopausal, or perimenopausal
- Histopathologically or cytologically confirmed ER+, HER2- breast cancer as per the ASCO/CAP guidelines
- ≥1 measurable lesion as per RECIST v1.1 or a mainly lytic bone lesion
- ECOG PS 0 or 1

Key Exclusion Criteria Phase 1b and Phase 2

- Active or newly diagnosed CNS metastases, including meningeal carcinomatosis
- Advanced, symptomatic visceral spread, that are at risk of life-threatening complications in the short-term
- Prior chemotherapy or elacestrant in the advanced/metastatic setting
- Concurrent malignancy or history of invasive malignancy within 3 years of enrollment
- Uncontrolled active infections
- Documented pneumonitis/ILD prior to Cycle 1 Day 1
- Major surgery within 28 days before starting trial therapy

METHODS – TREATMENT

Phase 1b: Dose Escalation

- Elacestrant dose escalation will be conducted using a standard 3+3 design. The starting dose of elacestrant dihydrochloride will be 300 mg when combined with alpelisib, everolimus, and palbociclib and 100 mg when combined with ribociclib.
- If 1 DLT is observed at any dose in all 3 cohorts, up to 3 more participants (for a total of 6 per dose level) will be treated at the same dose. If no additional DLTs are observed, participants may be enrolled in the next dose level.
- If ≥2 DLTs are observed in Cohort 1, the combination at the doses evaluated in Cohort 1 will be considered not feasible, and lower doses may be tested.
- If ≥2 DLTs are observed in Cohorts 2 or 3, the doses evaluated in Cohort 1 and 2 will be considered the RP2D of this combination, respectively.

Phase 2

- Patient enrollment, assignment of patients to specific cohorts/treatment arms, and selection of certain combinations of drugs in certain arms of Phase 2 will be selected by the investigator, based on the inclusion/exclusion criteria specific for each arm.

REFERENCES:
1. Lei JT, et al. *Breast*. 2019;48(Suppl 1): S26-S30.
2. Bidard FC, et al. *J Clin Oncol*. 2022;40:3246-3256.

ASSESSMENTS

- Tumor assessments will include:
 - Chest: CT
 - Abdomen, pelvis, and other known or suspected sites of the disease: CT or MRI
 - Brain: CT or MRI (brain assessment only required if the patient has history of brain metastases or symptoms suggestive of brain metastasis)
- Tumor assessments per RECIST v1.1 will be performed during screening/baseline period, every 8 weeks after cycle 1 day 1, and as indicated if disease progression is suspected clinically
- Safety assessments will consist of monitoring and recording all AEs, including all CTCAE v5.0 grades.
- Blood samples for PK will be collected for all patients enrolled in Phase 1b.
- Blood samples for PD and other biomarkers will be collected throughout the study.

ENDPOINTS

Primary

Phase 1b
- Number of DLTs observed during the first cycle

Phase 2
- Estimation of PFS rate at 6 months for each of the combination arms in patients who received prior AI and a CDK4/6i in the metastatic setting (Arms A, B, and C)
- Estimation of PFS rate at 12 months for all the combinations with palbociclib, abemaciclib, and ribociclib as second/third line treatment in the metastatic setting in patients who received no prior CDK4/6i in the metastatic setting (Arm D)

Secondary

Phase 1b
- AEs and SAEs, as well as changes in clinical laboratory values, vital sign measurements, and ECG parameters
- Standard plasma PK parameters
- ORR, DoR, CBR, PFS, OS

Phase 2
- ORR, DoR, CBR, PFS, and OS
- AEs and SAEs, as well as changes in clinical laboratory values, vital signs, and ECG parameters

Exploratory

Phase 1b
- Changes in biomarkers including, but not limited to, allele mutation frequencies in cfNA in blood
- Relationship between efficacy endpoints and allele mutation frequencies (at baseline and post-baseline)
- ORR, DoR, CBR, PFS, and OS

Phase 2
- Changes in biomarkers including, but not limited to, allele mutation frequencies in cfNA in blood
- Relationship between efficacy endpoints and allele mutation frequencies (at baseline and post-baseline)
- ORR, DoR, CBR, PFS, and OS

AI, aromotase intibitor; cfNA, cell-free nucleic acid; DLT, dose-limiting toxicity; DoR, duration of response; ECG, electrocardiogram; ORR, overall response rate; PK, pharmacokinetics; OS, overall survival; SAE, serious adverse event

- For updated trial information, please visit clinicaltrials.gov NCT05563220 or contact ELEVATE@stemline.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster.